MTG does not increase immuno-detectable amounts of deamidated gliadin at standard bakery concentrations

Andreas Heil[1], **Jürgen Ohsam**[1], **Bernard van Genugten**[2], **Oscar Diez**[2], **Keiichi Yokoyama**[3], **Yoshiyuki Kumazawa**[3], **Ralf Pasternack**[1], **Martin Hils**[1]

[1]Zedira GmbH, Darmstadt, Germany, [2]AB Enzymes GmbH, Darmstadt, Germany, [3]Ajinomoto Co., Inc., Institute of Food Sciences & Technologies, Kawasaki-Shi, Japan

Introduction and Objective

Microbial transglutaminase (MTG) is an enzyme derived from *S. mobaraensis* which catalyzes cross-linking of protein-bound glutamine to protein-bound lysine. The cross-linking function is used to improve properties of protein-containing food like meat, sausage, fish, dairy products, pasta and bread. The main technological benefits of MTG in bakery applications is the positive effect on dough stability directly after mixing. At high concentrations, the loaf volume is reduced as a result of a strong meshwork of cross-linked proteins and therefore restricted expansion of gas bubbles.

Typically, MTG is never used as a single enzyme in a given bakery application but always in combination with other technologically required enzyme classes. It can be considered a niche enzyme within the group of typical baking enzymes, such as amylases, xylanases, etc., and the global market share of MTG used for baking is estimated to be lower than 1% of the total baking enzyme market. This low share is a result of its relatively low dosage (VERON TG is used at 1–3 g/100 kg of flour, corresponding to 1–3 units/kg of flour). MTG is sold and used for bakery applications on all continents, without any significant focus countries. It is only required for solving explicit technical cases, such as industrial bakers using wheat dough with short proofing times.

Besides cross-linking, MTG is able to deamidate gluten peptides under certain conditions (Heil et al., 2016). In celiac disease (CD), gluten deamidation by human tissue transglutaminase (TG2) is a key step in pathogenesis (Molberg et al., 1997).

As a result of this analogy, since about a decade, concerns are raised regarding the use of MTG in gluten-containing food products because of the potential generation of CD-specific deamidated immunogenic epitopes (Gerrard et al., 2005). Therefore, the aim of this study was to investigate the effect of common industrially applied MTG concentrations in wheat bread production on the formation of deamidated gluten epitopes.

Methods and Results

Analysis of wheat gluten extracts from MTG-treated breads

Albumin/globulin, gliadin and glutenin extracts were analyzed using SDS-PAGE and Western blotting with an antibody detecting unmodified gliadin (A011) (Fig. 1). Similar protein amounts were extractable for each protein fraction of control (0), 2, 4 and 8 U MTG/kg bread (Fig. 1 A, B, C). Western blot analysis revealed only little gliadin detectable in the aqueous albumin/globulin extracts. More intensive staining was observed for gliadin and glutenin extracts from the samples (Fig. 1 D, E, F).

No differences in the band pattern between the 0, 2, 4 and 8 U MTG/kg samples were observed, neither in silver stained gels nor in western blotting. Further, no influence of MTG on the protein composition of the bread samples prepared with increasing amounts of MTG could be detected. In the positive control extract prepared with the extraordinarily high MTG-concentration of 2,000 U MTG/kg, high molecular weight bands are present predominantly for gliadins and glutenins.

Analysis of MTG-treated bread for gliadin deamidation

A monoclonal antibody (A057) raised against deamidated alpha2-gliadin 33mer (Skovbjerg et al, 2004) was used to detect possible deamidation of gluten by MTG. Western blot analysis showed that in the industrially prepared bread samples (0, 2, 4 and 8 U MTG/kg flour), basically no staining was observed (Fig. 2). However, for the 2,000 U MTG/kg control, staining of multiple bands with a molecular size of 36-40 kDa was observed in every extract.

Analysis of MTG-treated bread using CD patient sera pool

A CD patient sera pool with a high titer of antibodies against deamidated gliadin peptides (DGP) was used for Western blot analysis (Fig. 3). In the albumin/globulin fractions, staining was absent. In the gliadin fractions a very faint signal was obtained for IgG-antibodies, but not for IgA-antibodies. In contrast, the 2,000 U MTG/kg control bread extracts as well as the DGPx2 controls showed a clear signal for both antibody classes. The glutenin extracts showed a clear staining but no difference in the band pattern between MTG-treated bread and control bread was observable.

While for controls DGPx2, gamma-gliadin 26mer and 2,000 U MTG/kg flour (gliadin fraction) a clear signal could be obtained, no staining was observed for the bread samples prepared with the low MTG amounts used for bakery products.

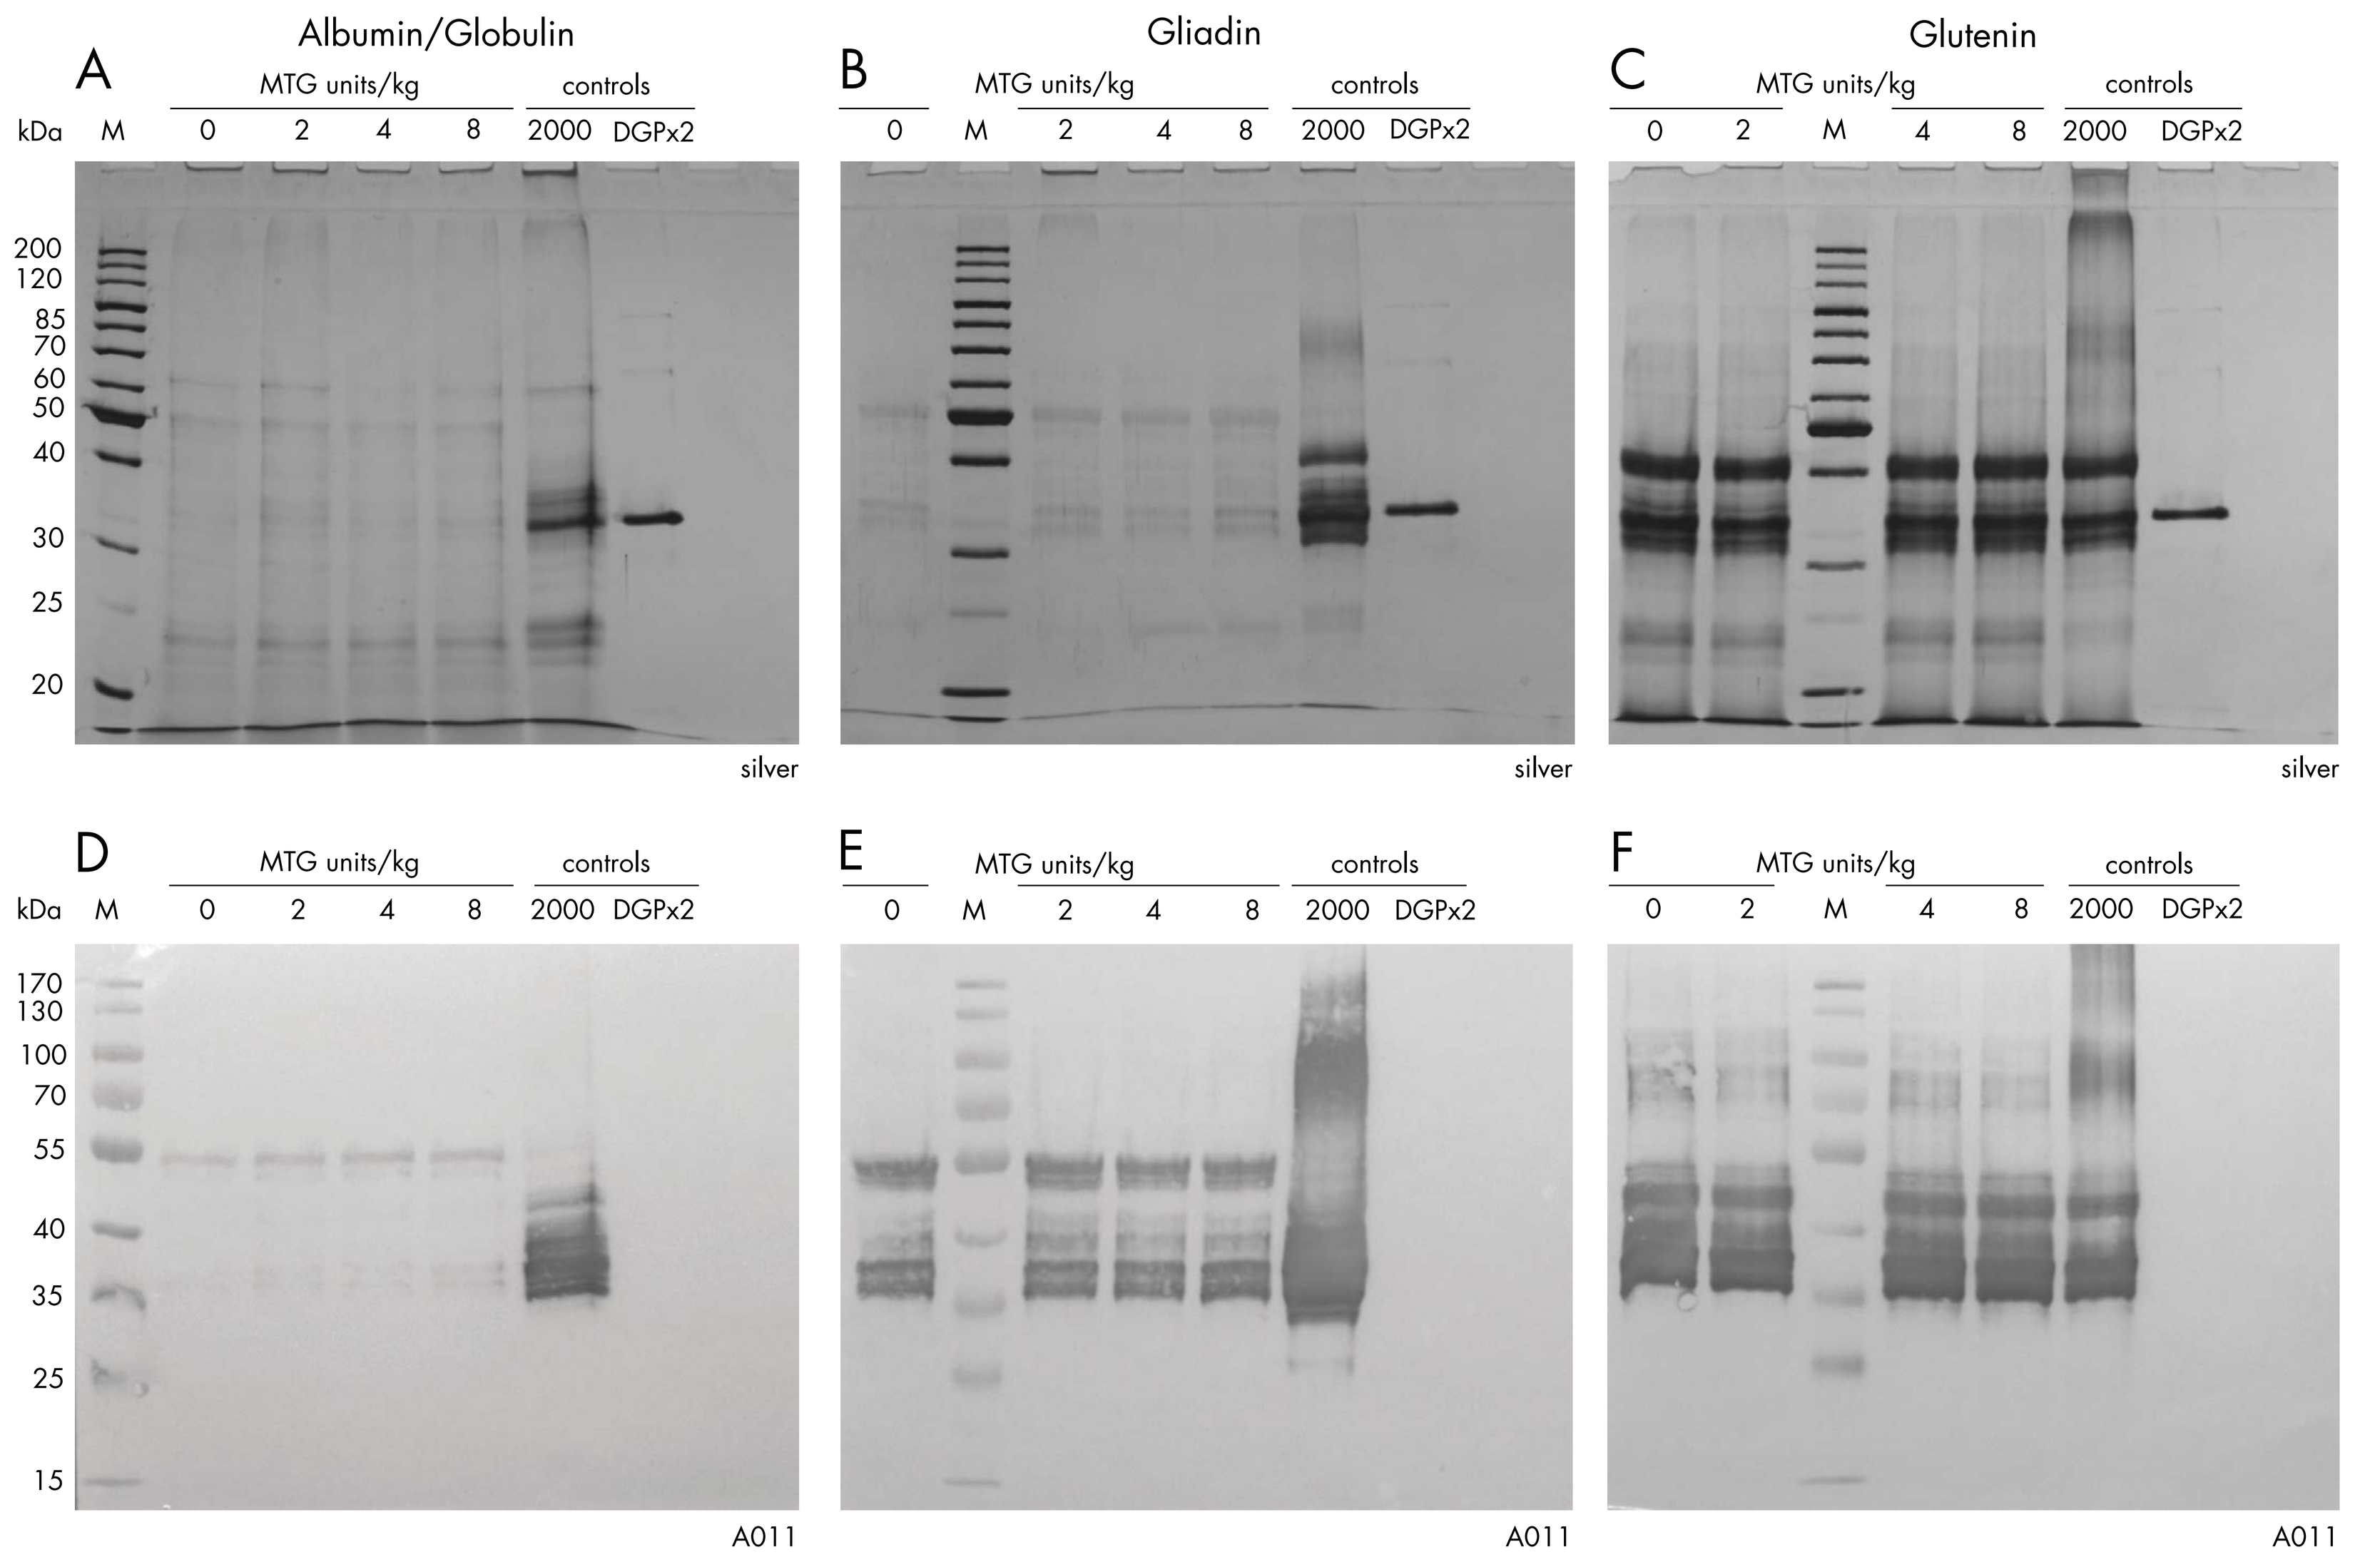
A
Albumin/Globulin
MTG units/kg
controls
kDa M 0 2 4 8 2000 DGPx2
B
Gliadin
0 M 2 4 8 2000 DGPx2
C
Glutenin
0 2 M 4 8 2000 DGPx2
200
120
85
70
60
50
40
30
25
20
silver
D
E
F
170
130
100
70
55
40
35
25
15
A011

Figure 1: SDS-PAGE and Western blot analysis of wheat protein fractions extracted from bread prepared without (0), 2, 4 and 8 U MTG/kg flour. A, B, C: silver-stained gels; D, E, F: Western blots using monoclonal antibody A011 specific to unmodified gliadin. Gliadin extract from 2,000 U MTG/kg flour and DGPx2 were deamidated gliadin controls.

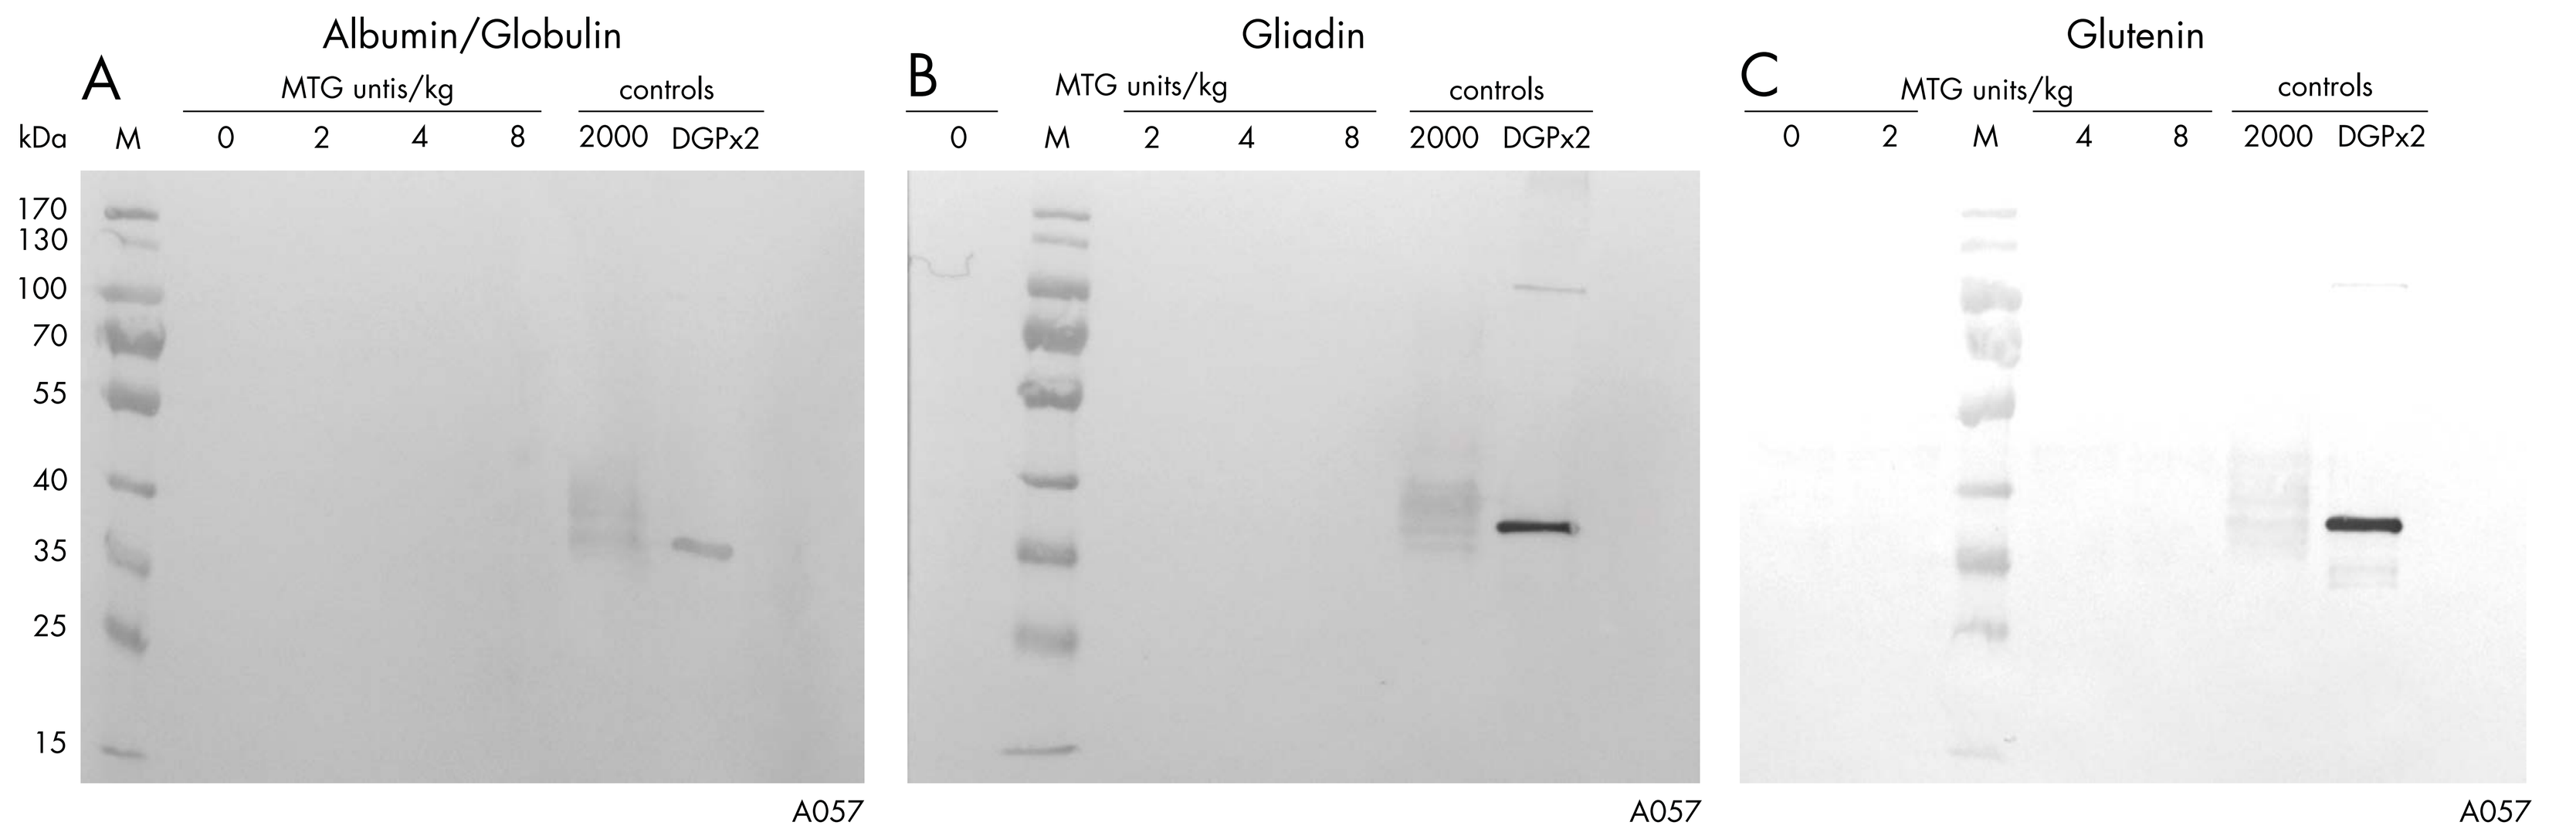
A
Albumin/Globulin
MTG untis/kg
controls
kDa M 0 2 4 8 2000 DGPx2
B
Gliadin
MTG units/kg
0 M 2 4 8 2000 DGPx2
C
Glutenin
0 2 M 4 8 2000 DGPx2
170
130
100
70
55
40
35
25
15
A057

Figure 2: Western blot analysis of extracts from MTG treated bread samples using deamidated gliadin specific antibody A057. DGPx2: deamidated gliadin control.

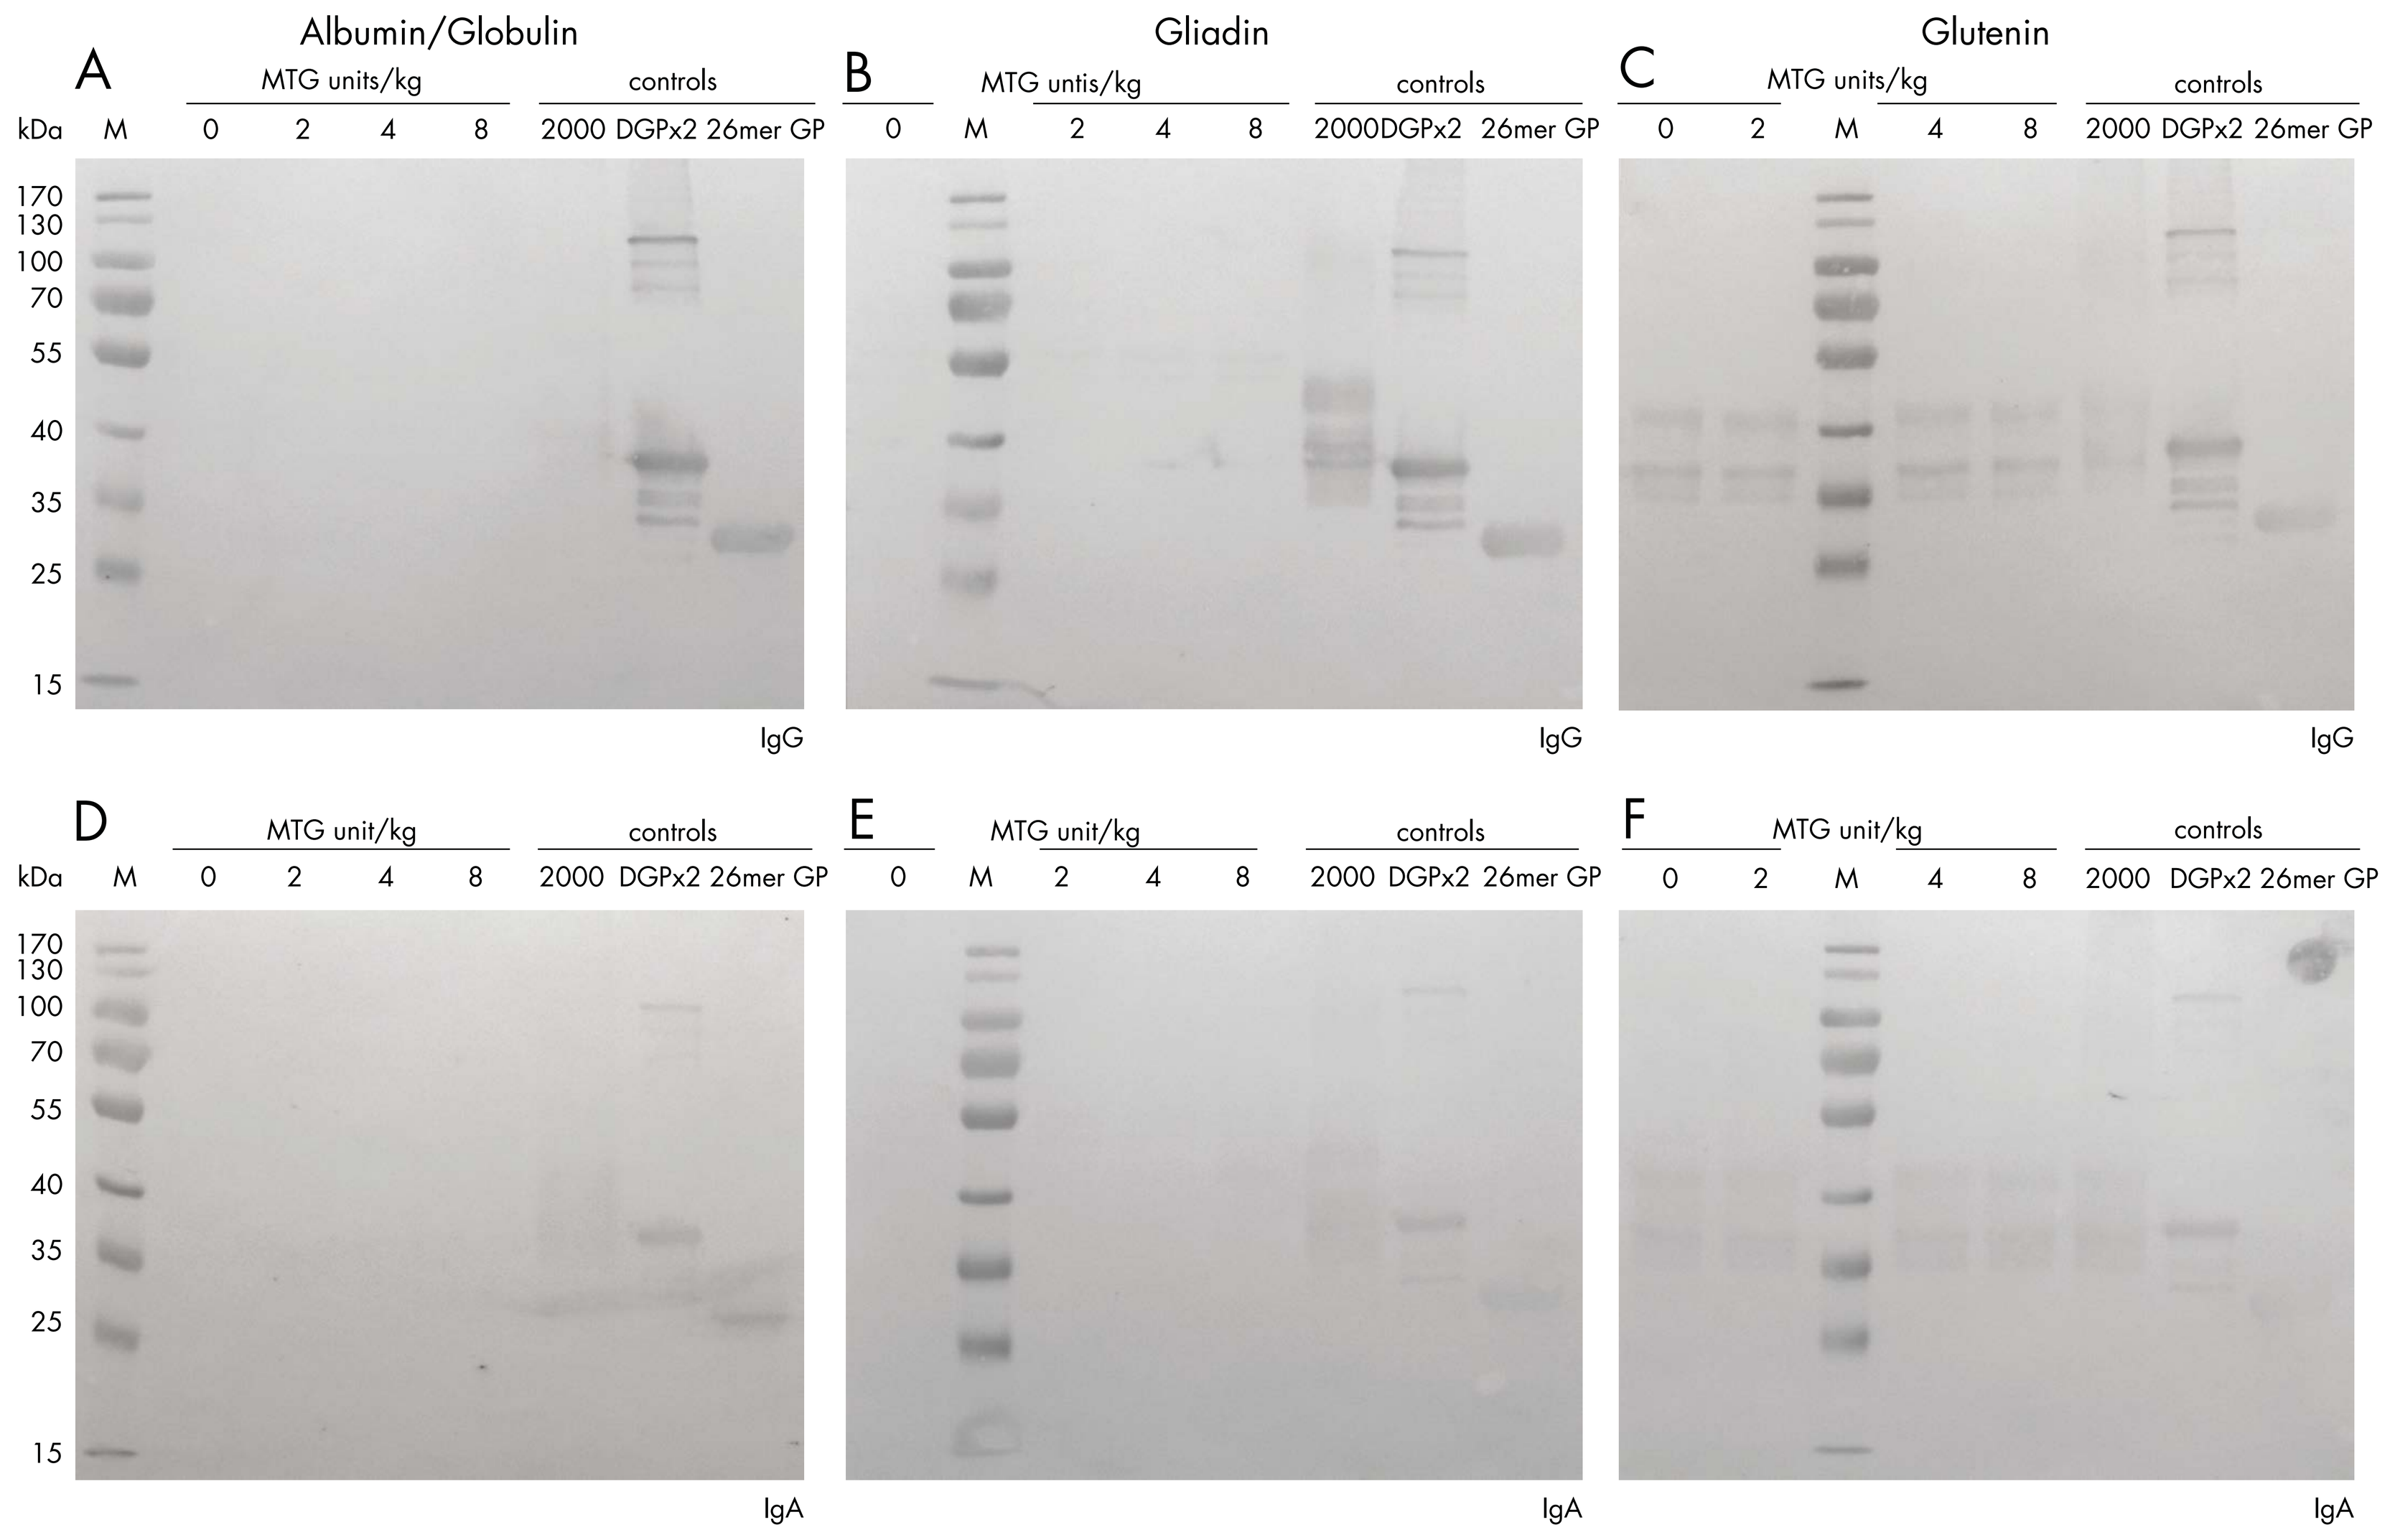
A
Albumin/Globulin
MTG units/kg
controls
kDa M 0 2 4 8 2000 DGPx2 26mer GP
B
Gliadin
0 M 2 4 8 2000DGPx2 26mer GP
C
Glutenin
0 2 M 4 8 2000 DGPx2 26mer GP
IgG
D
MTG unit/kg
E
F
IgA
170
130
100
70
55
40
35
25
15

Figure 3: Western blot analysis of MTG treated bread samples using celiac disease patients' sera. A, B, C: Detection of IgG antibodies; D, E, F: Detection of IgA antibodies. DGPx2: deamidated gliadin control. 26mer GP: non-deamidated gliadin control.

Conclusion

Our results indicate that MTG-treatment of wheat bread prepared with MTG concentrations typically used in standard bakery processes does not lead to immuno-detectable amounts of CD-immunotoxic deamidated gliadins.

References

Molberg et al., Nature Medicine, 1997

Heil et al., Journal of Cereal Science, 2016

Gerrard et al., Trends in Food Science and Technology, 2005

Skovbjerg et al., Biochimica et Biophysica Acta, 2004

© 2017, Zedira GmbH